भारत की पोर्टुनिड केकड़ा संपदाएं
PORTUNID CRAB RESOURCES OF INDIA

Infraorder Brachyura Linnaeus, 1758
Superfamily Portunoidea Rafinesque, 1815

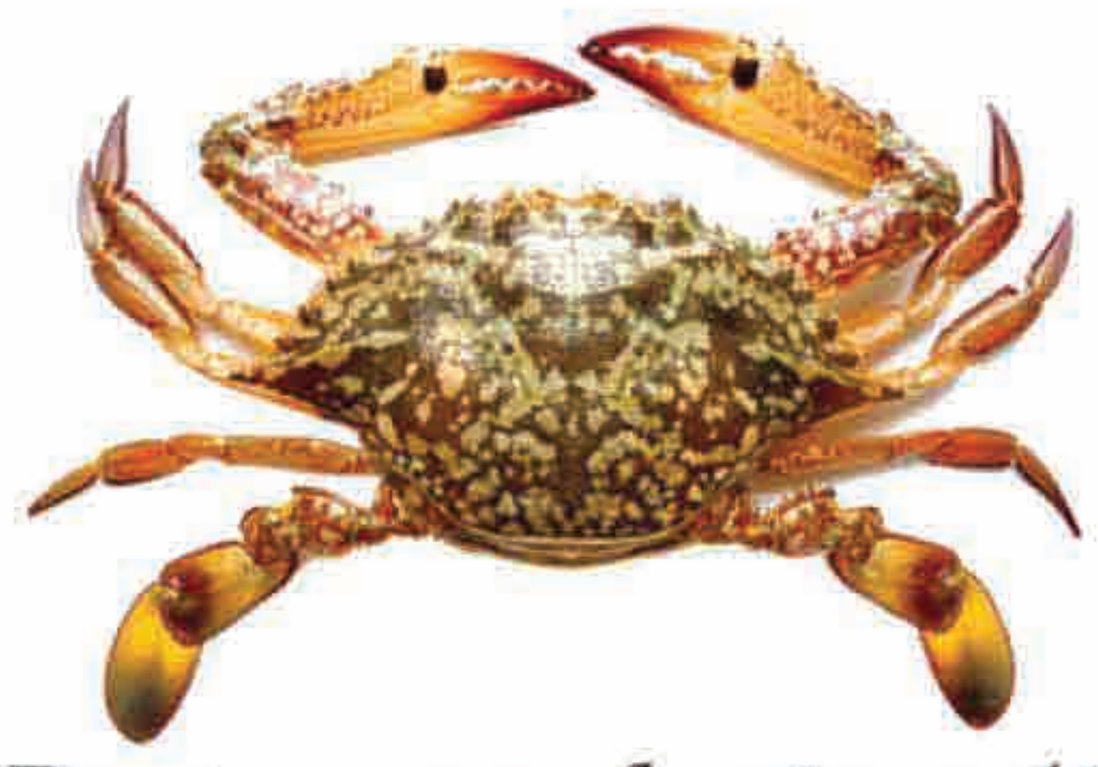

Portunus pelagicus (♀)
(Linnaeus, 1758)
Flower crab

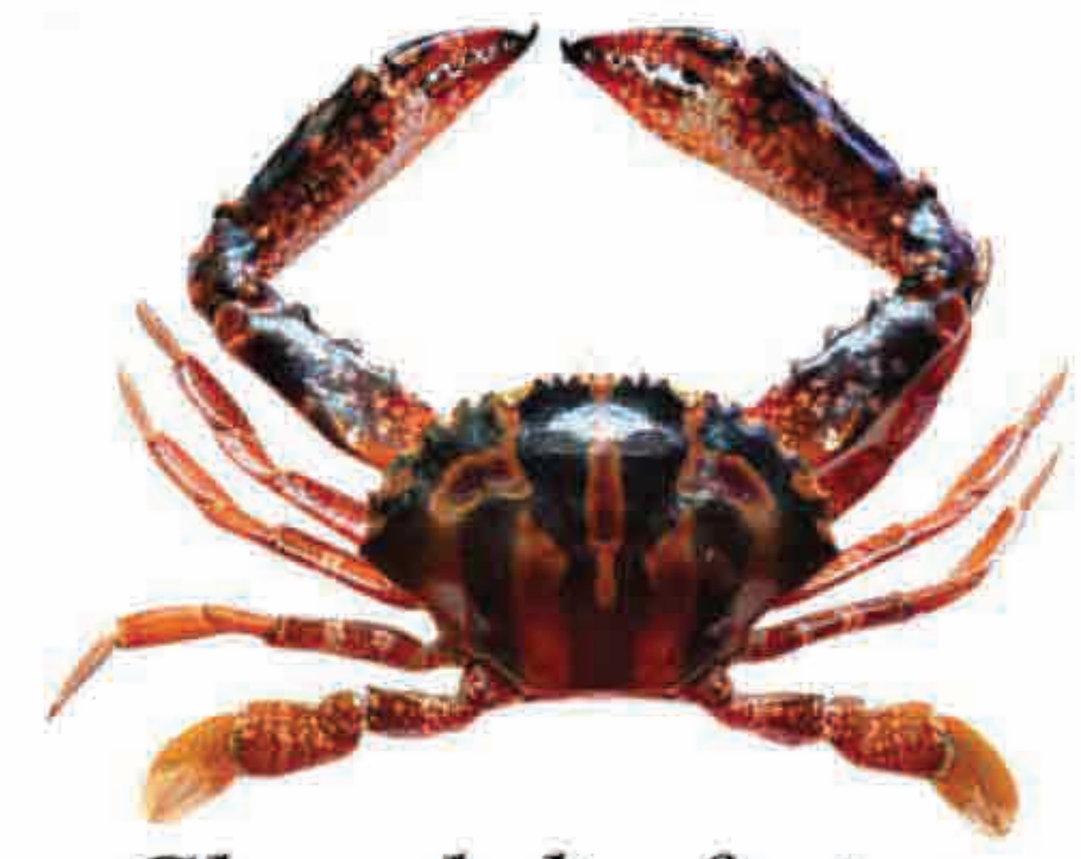

Charybdis feriatus
(Linnaeus, 1758)
Crucifix crab

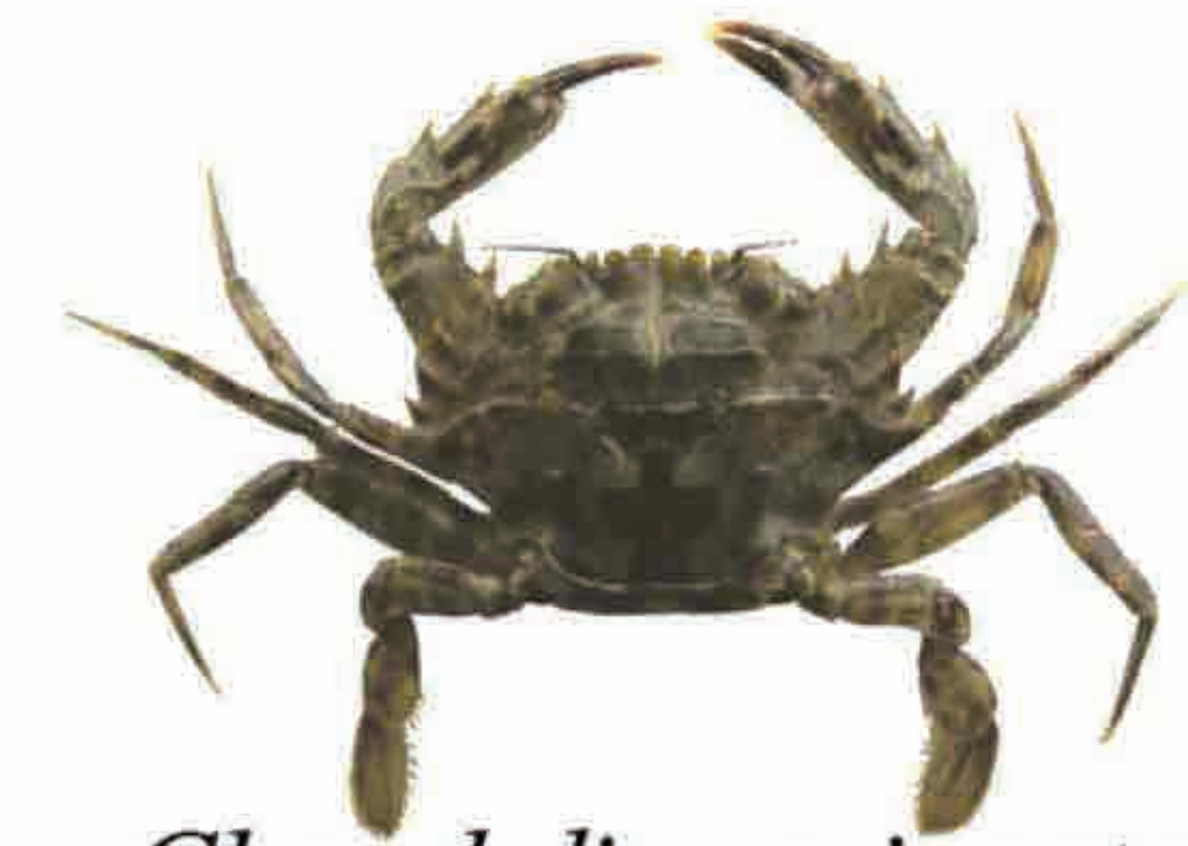

Charybdis variegata
(Fabricius, 1798)

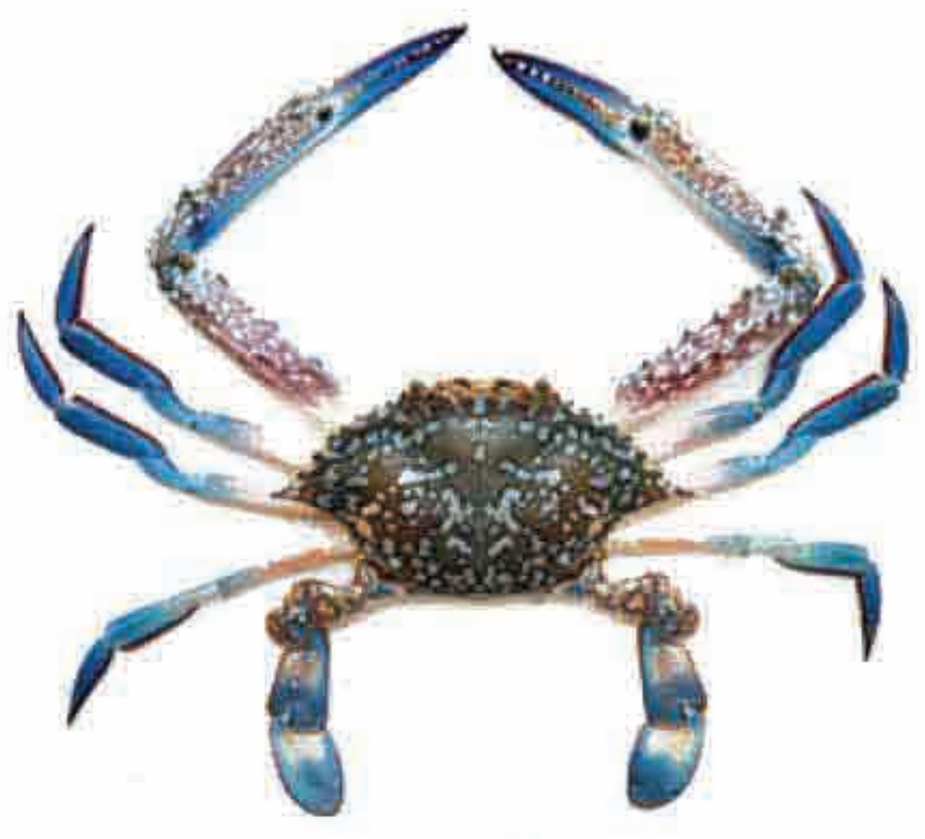

Portunus pelagicus (♂)
(Linnaeus, 1758)
Flower crab

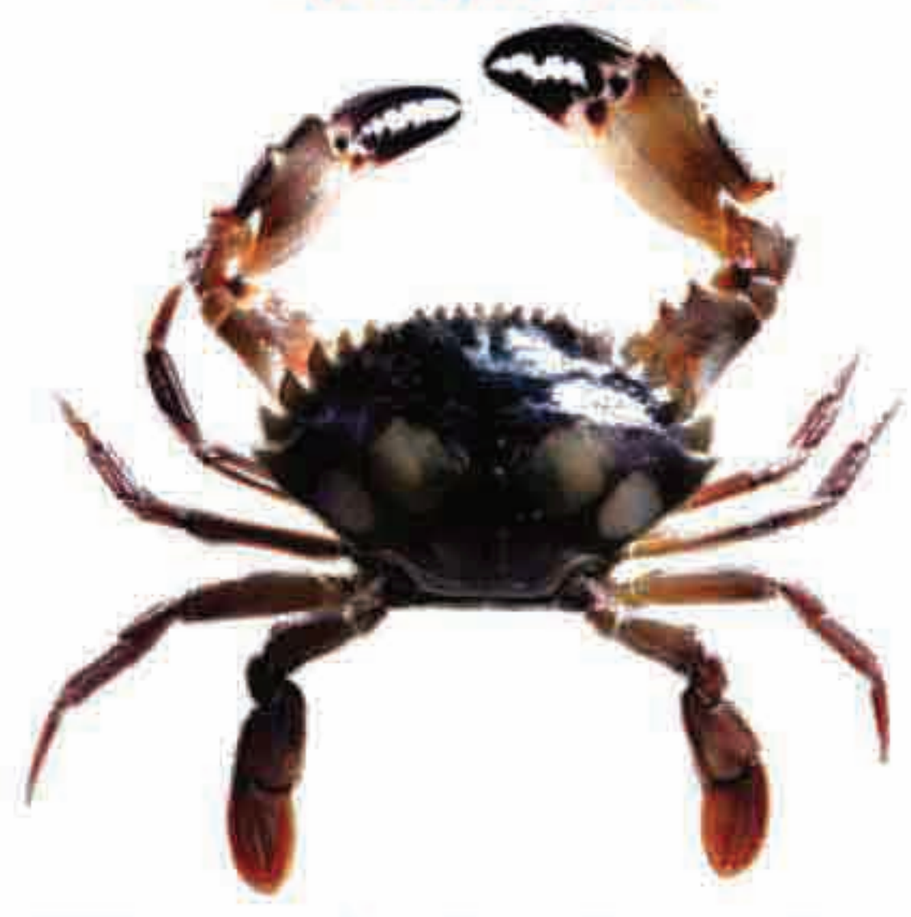

Charybdis lucifera
(Fabricius, 1798)
Yellow brown crab

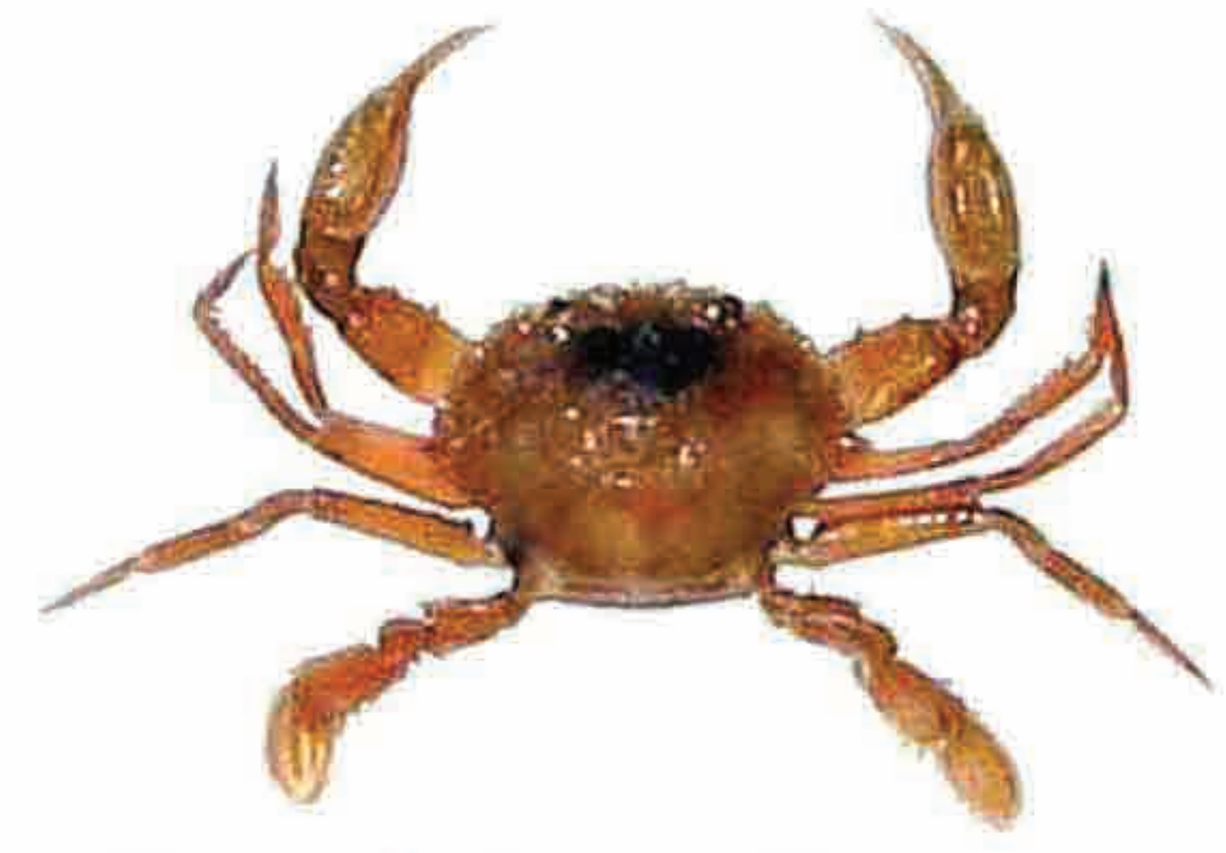

Charybdis callianassa
(Herbst, 1789)

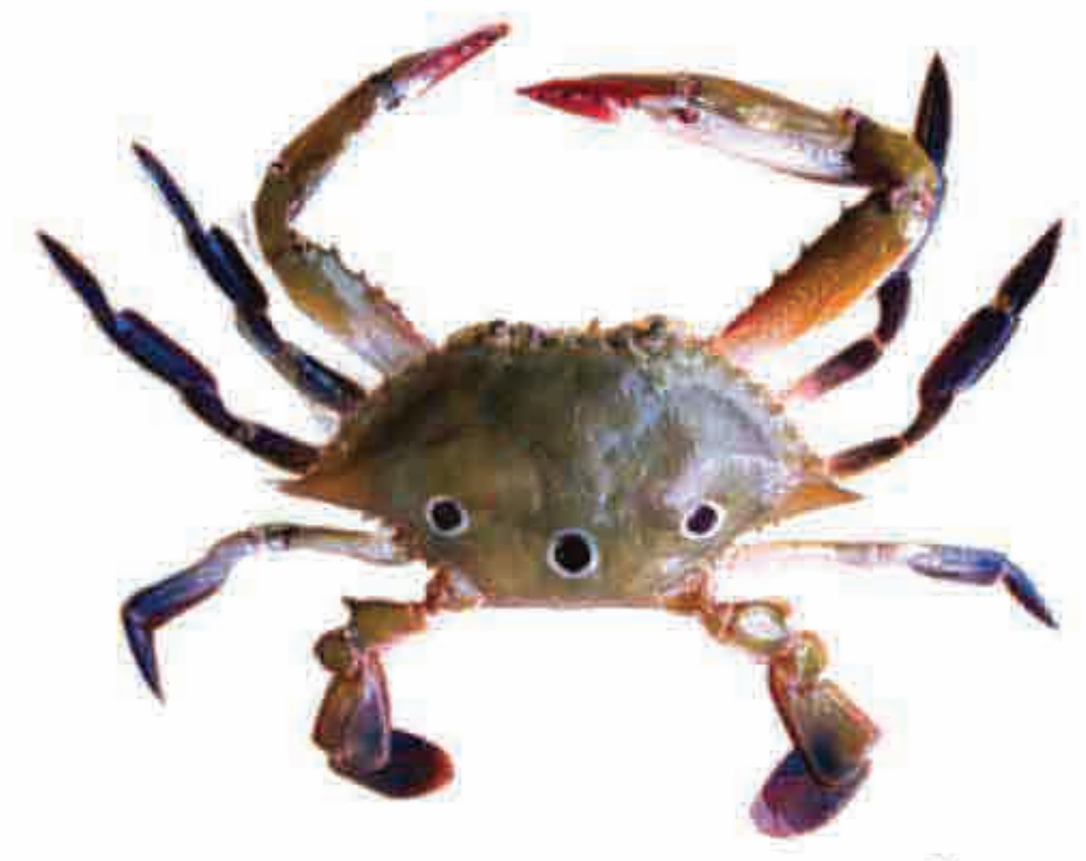

Portunus sanguinolentus
(Herbst, 1783)
Blood Spotted crab

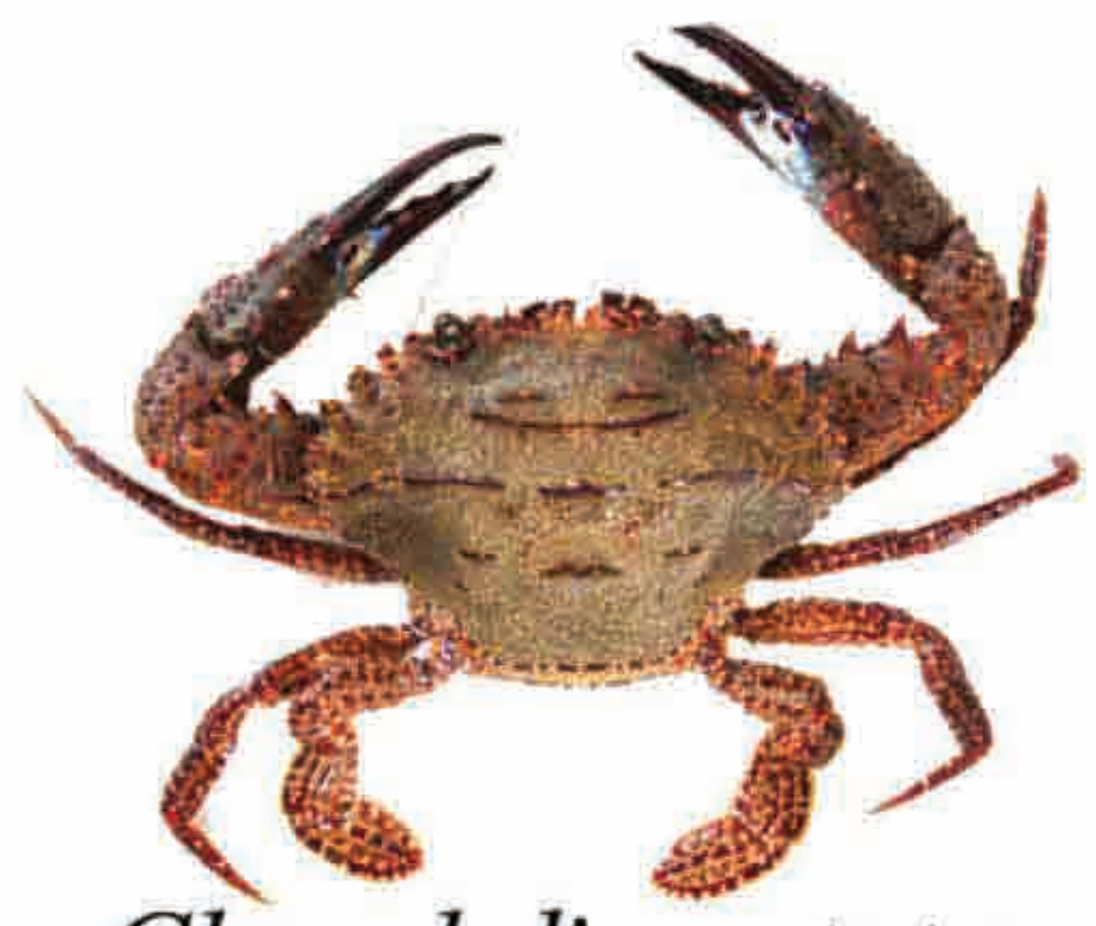

Charybdis natator
(Herbst, 1794)
Ridged swimming crab

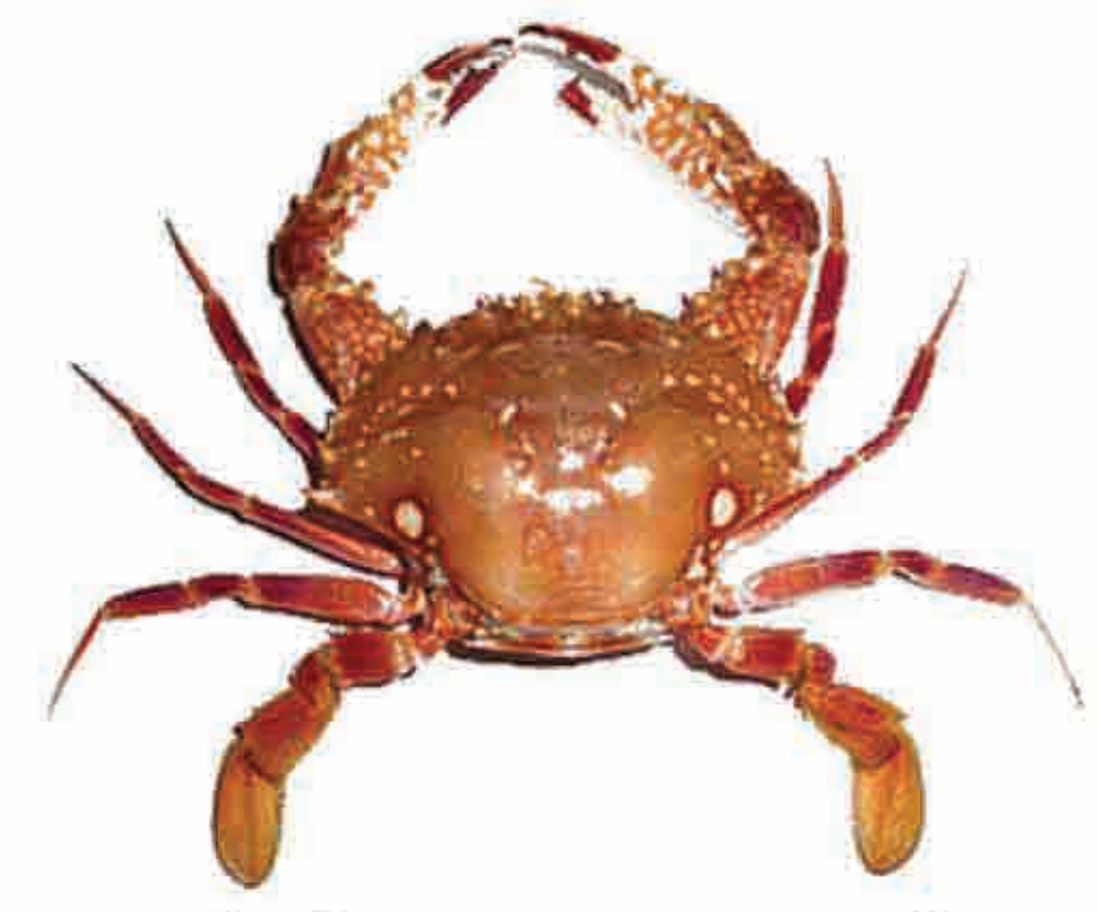

Charybdis riversandersoni
Alcock, 1899

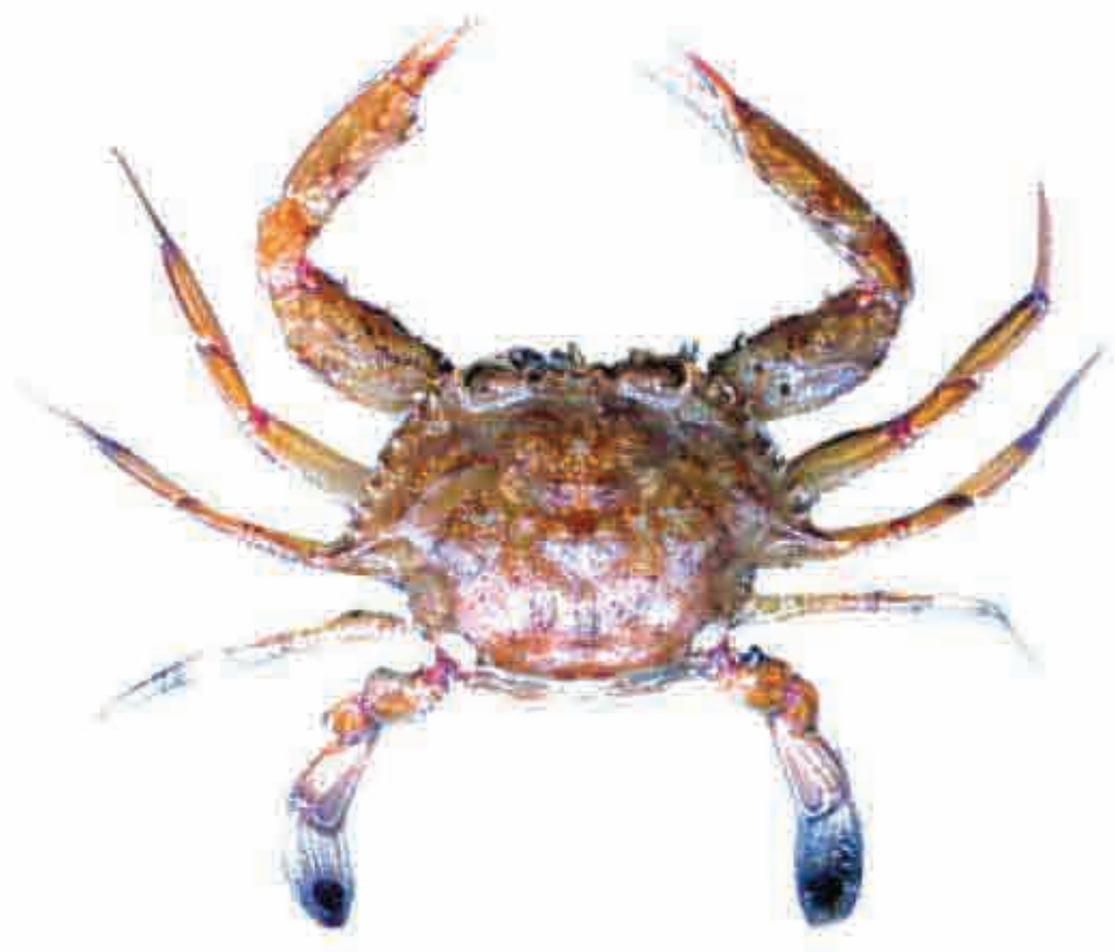

Portunus argentatus
(A. Milne Edwards, 1861)
Warty swimming crab

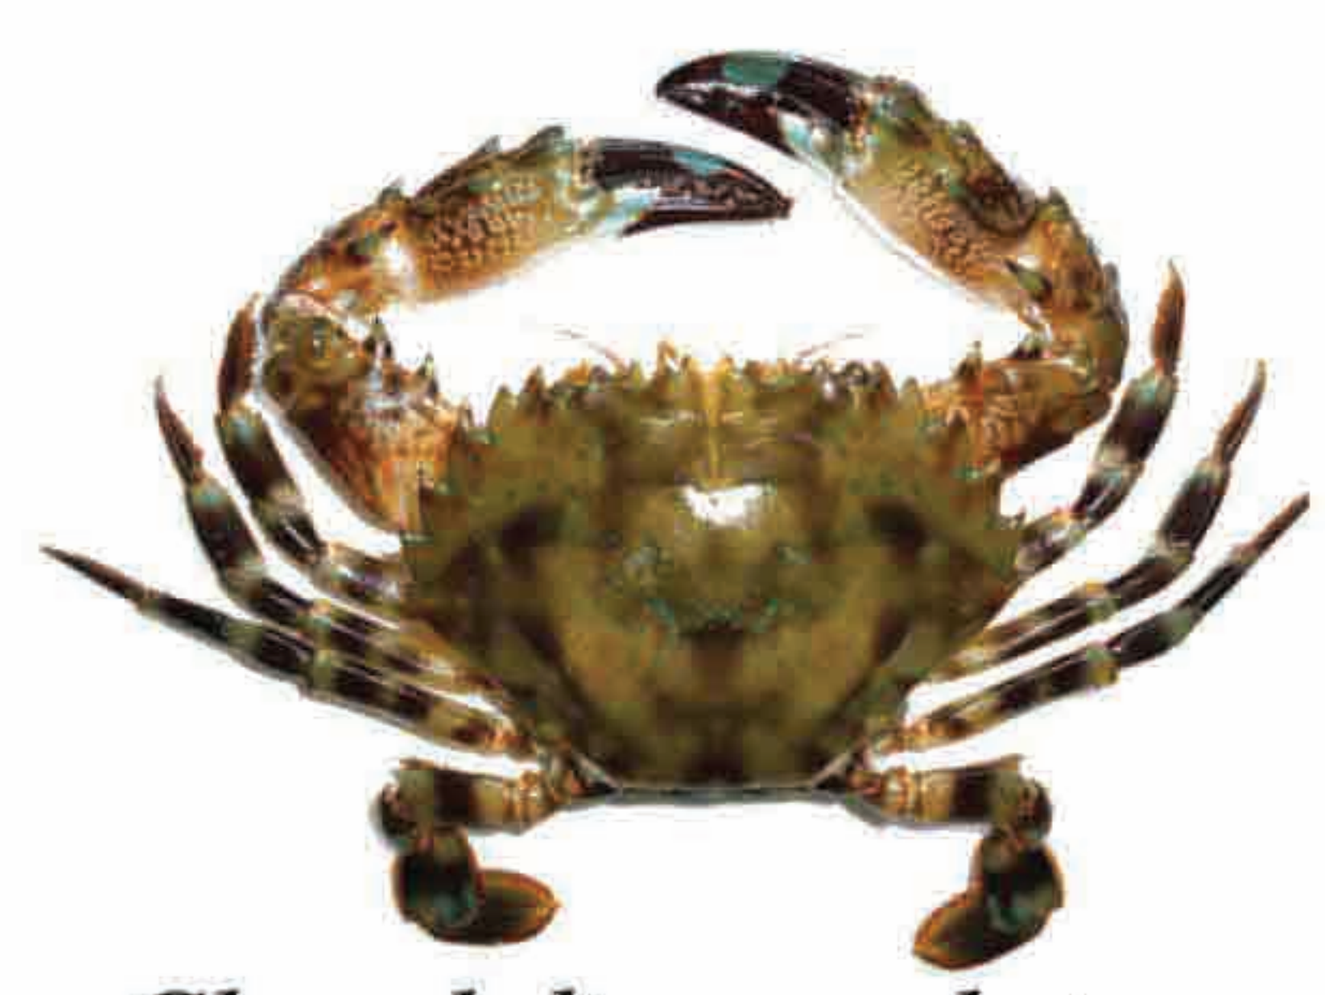

Charybdis annulata
(Fabricius, 1798)
Banded leg swimming crab

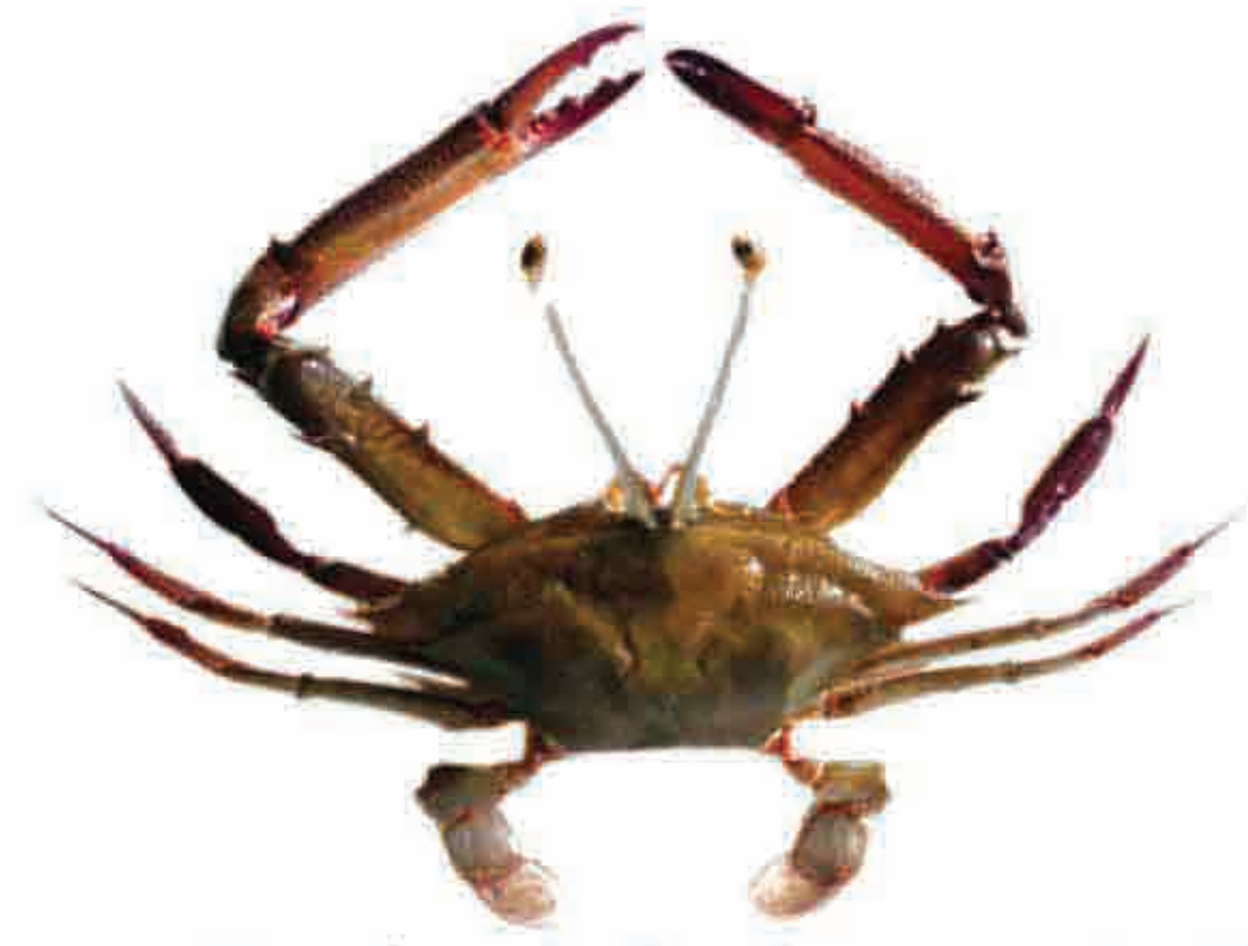

Podophthalmus vigil
(Fabricius, 1798)
Sentinel crab

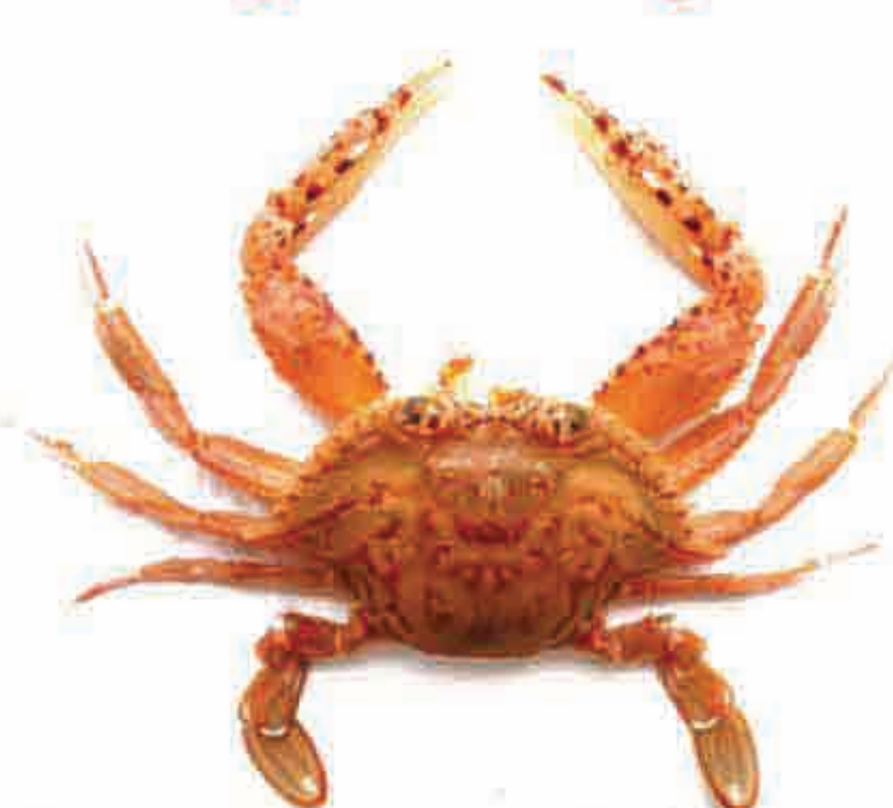

Portunus gladiator
Fabricius, 1798

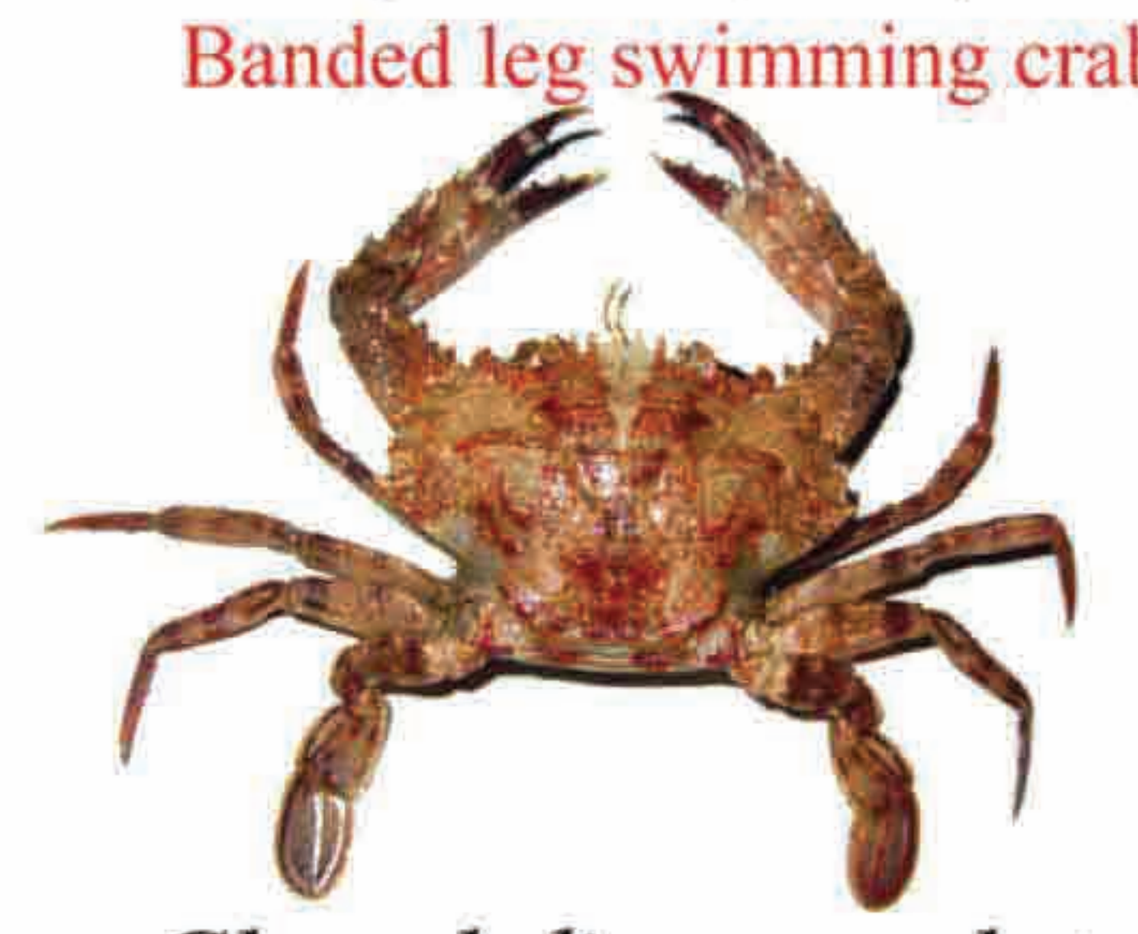

Charybdis granulata
(De Haan, 1833)

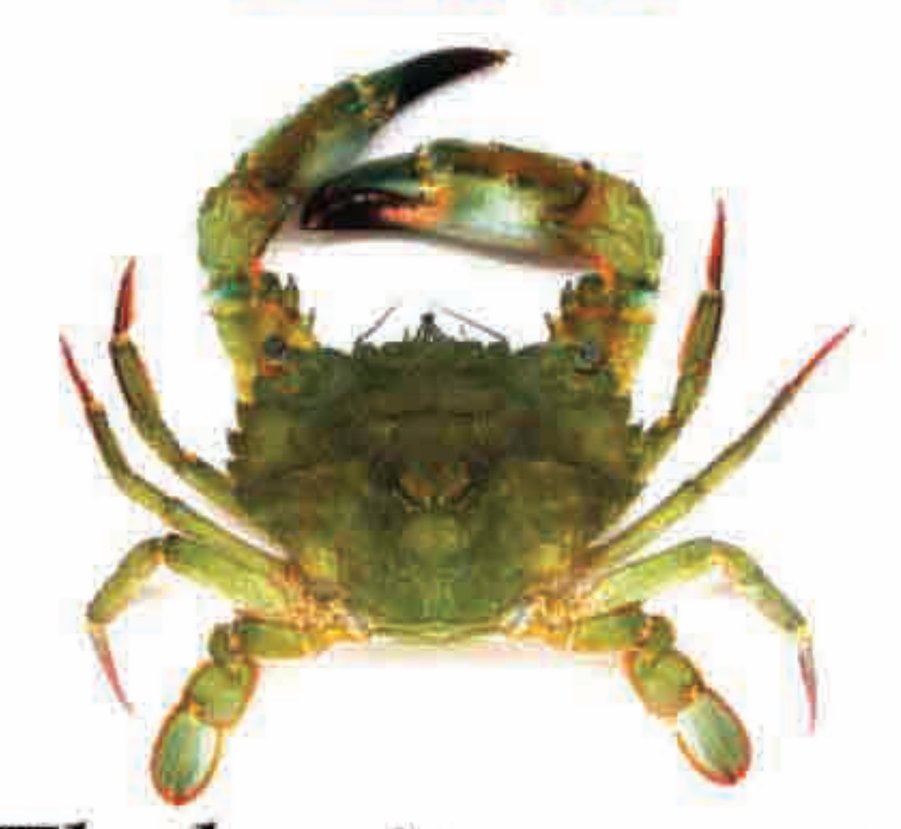

Thalamita crenata
Rupell, 1830
Crenata swimming crab

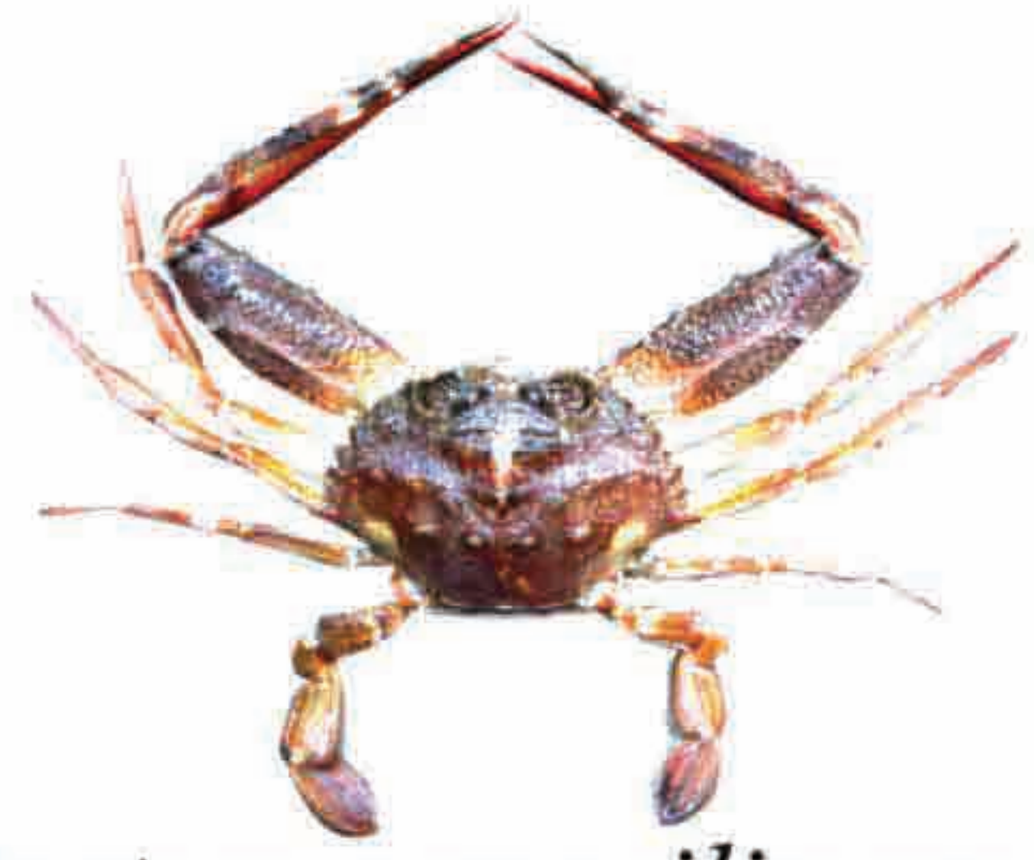

Portunus gracilimanus
(Stimpson, 1858)

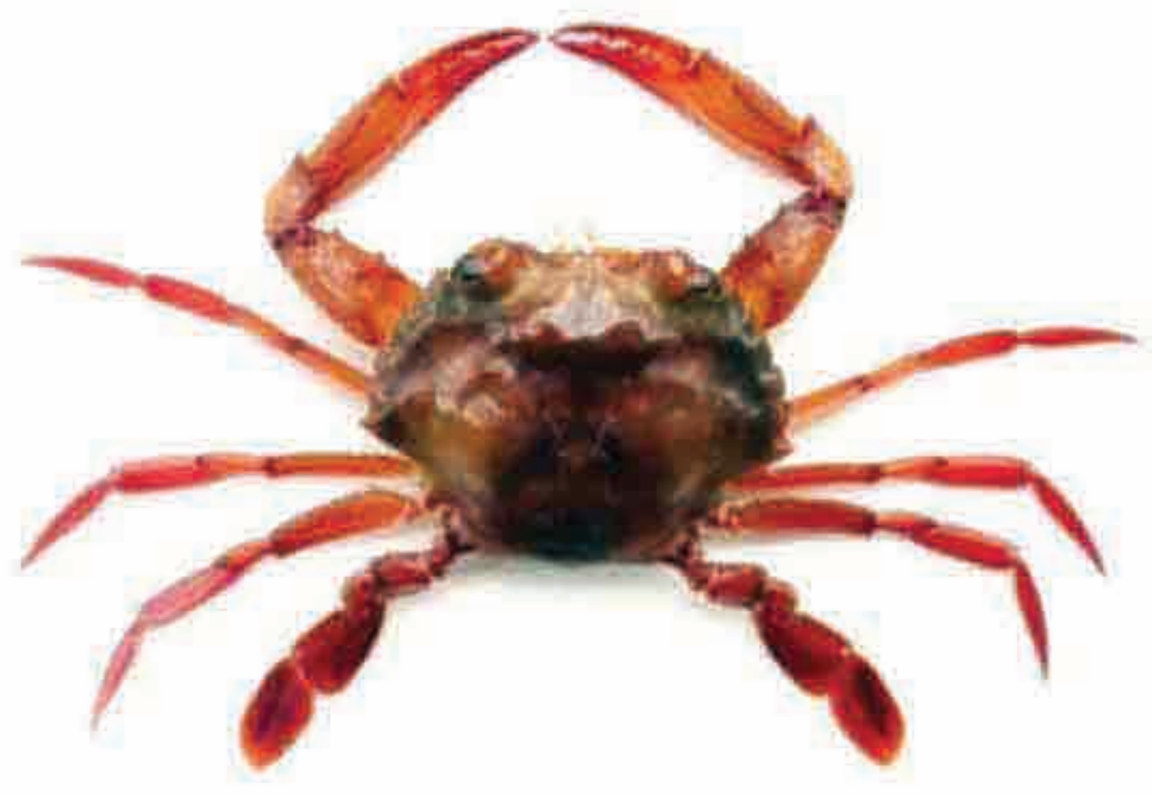

Charybdis smithii
MacLeay, 1838
Swarming crab

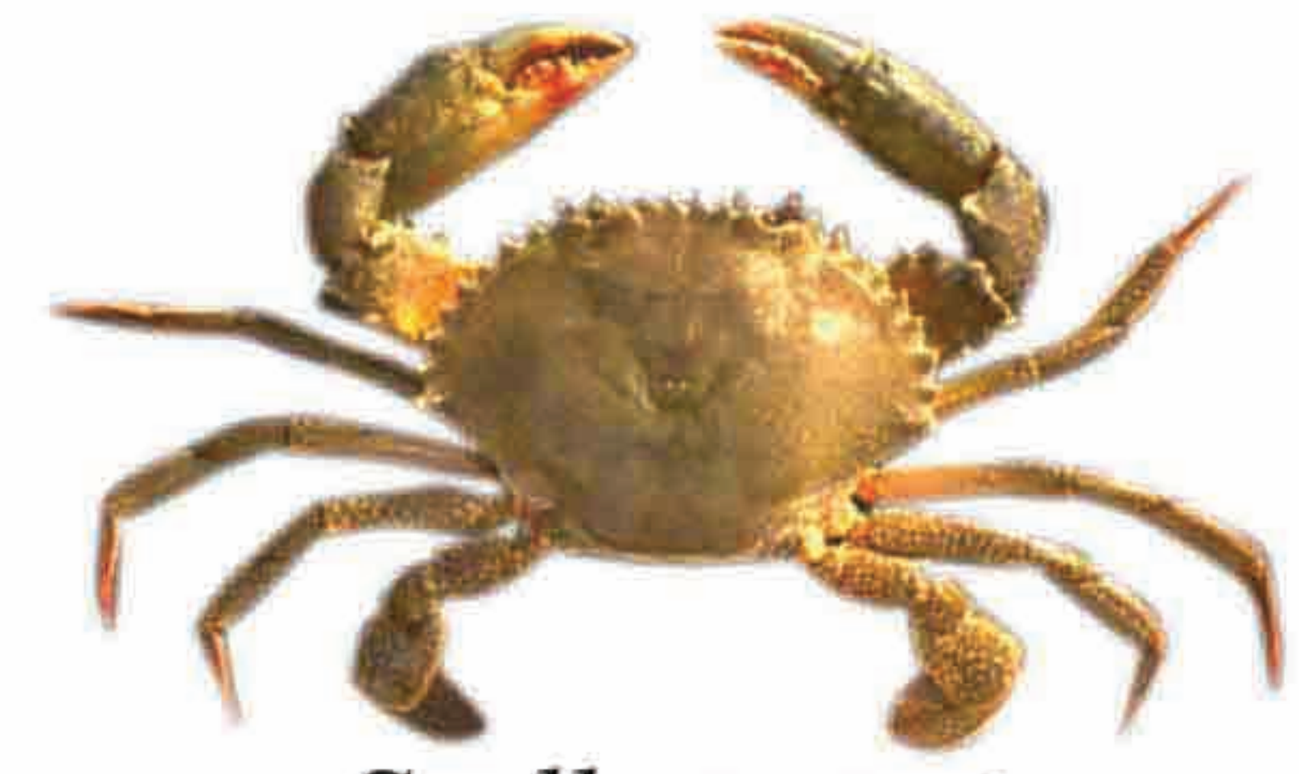

Scylla serrata
(Forsskal, 1775)
Giant mud crab

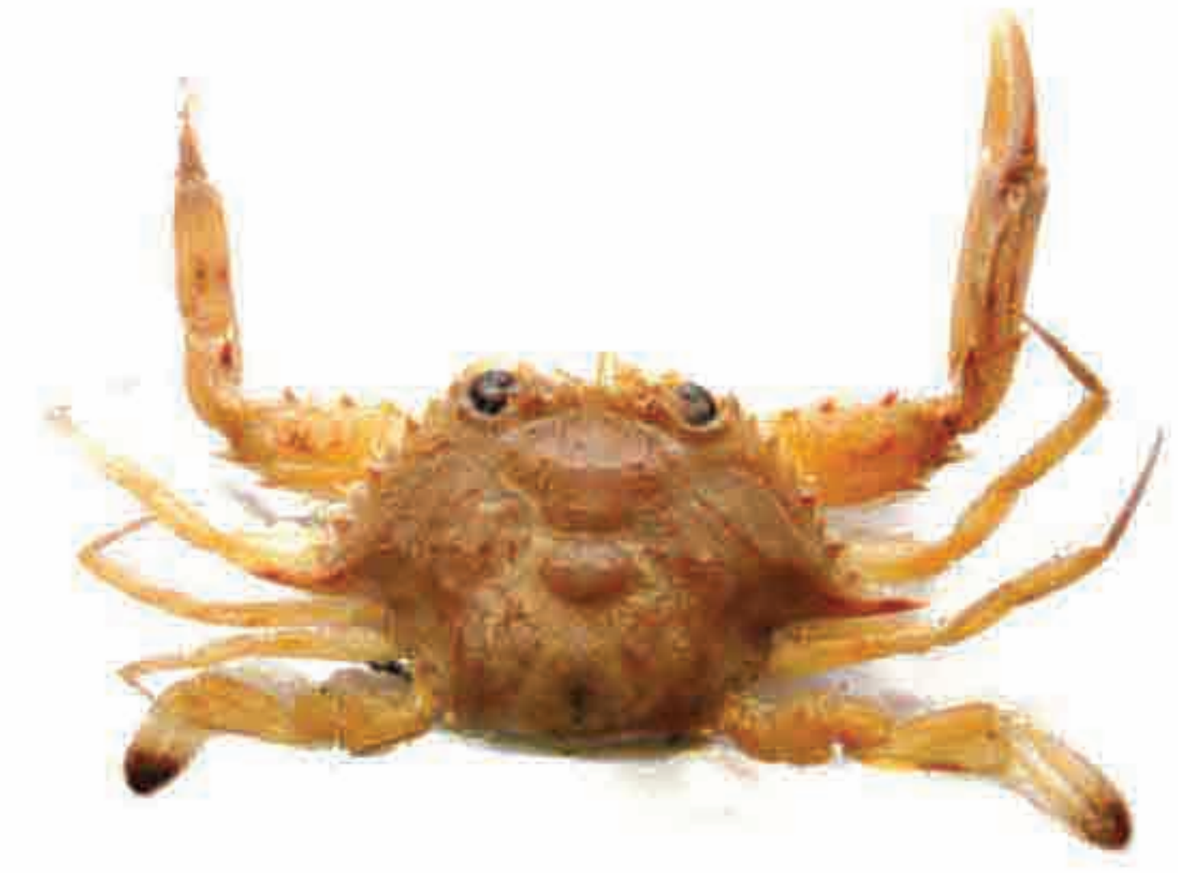

Portunus hastatoides
Fabricius, 1798

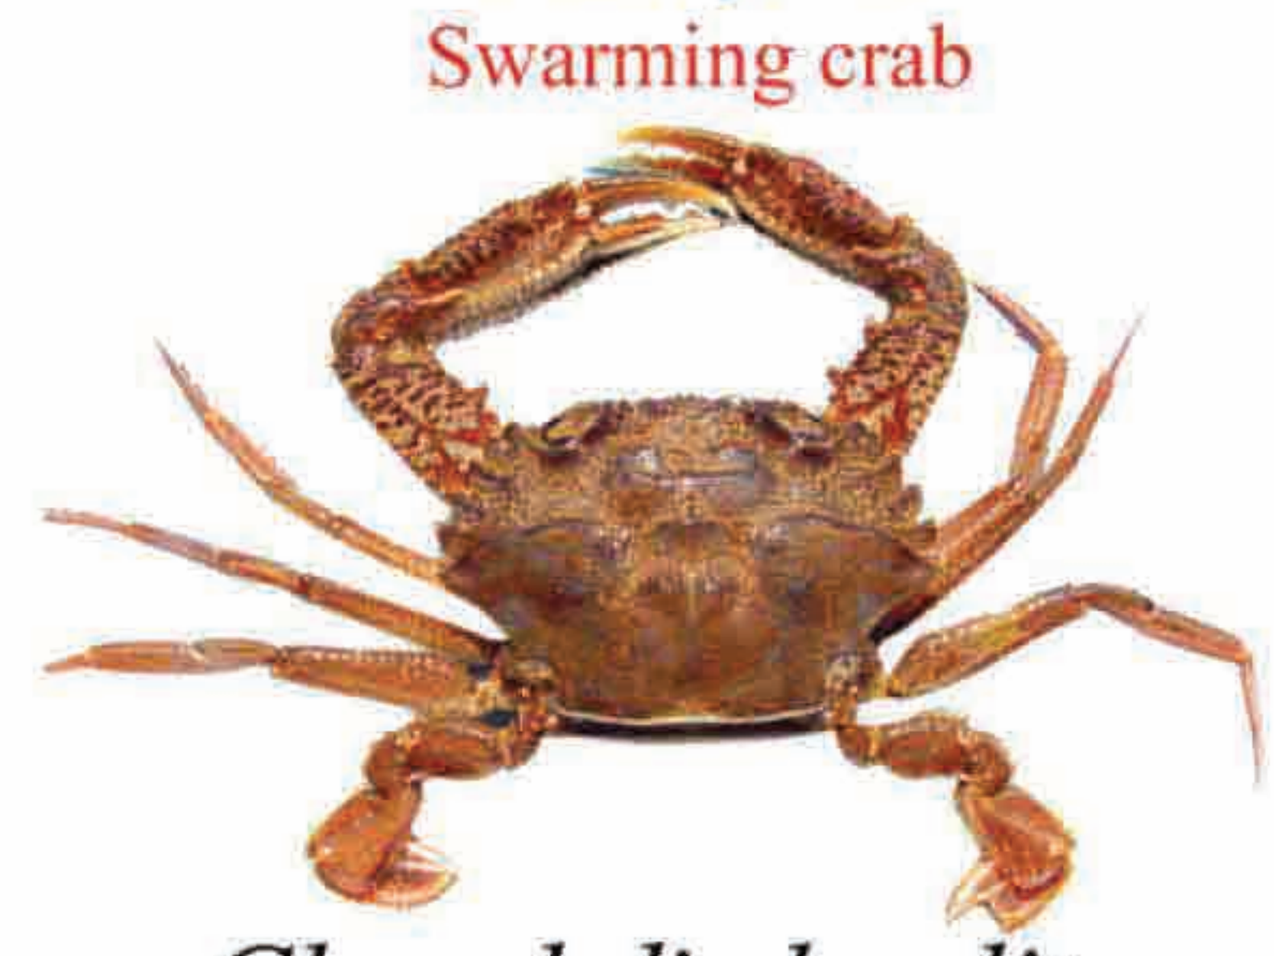

Charybdis hoplites
(Wood- Mason, 1877)

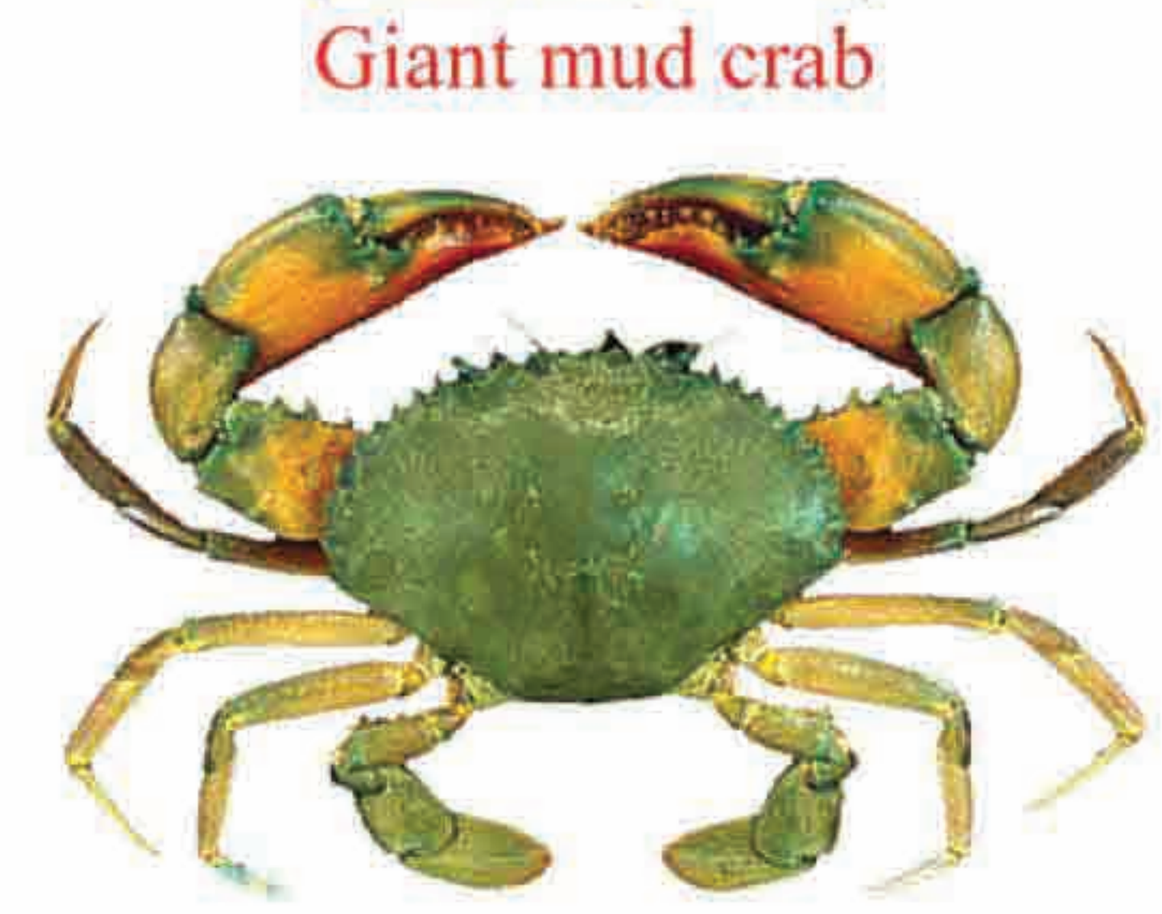

Scylla olivacea
(Herbst, 1796)
Orange mud crab

तैयारीः एस. लक्ष्मी पिल्लै, जोसलीन जोस, जी.महेश्वरुडू, पी. तिरुमिलू, पी.के. बेबी और एम. राधाकृष्णन
Prepared by : S. Lakshmi Pillai, Josileen Jose, G. Maheswarudu, P. Thirumilu, P.K. Baby & M. Radhakrishnan
प्रकाशनः निदेशक, केंद्रीय समुद्री मात्स्यिकी अनुसंधान संस्थान, कोचीन - 682018, केरल, भारत.
Published By: Director, Central Marine Fisheries Research Institute, Kochi - 682018, Kerala, India.

CMFRI Poster No. 5/2014